[APS/Physiology, Abstract R2343 (program #586.1)]

Probiotic Effect on SARS-CoV2 Immunity Is Associated to Type-1 Interferons: A Post-Hoc Analysis of a Randomized, Placebo-Controlled Trial

Pedro Gutiérrez-Castrellon[1], Tania Gandara-Marti[1] and Jordi Espadaler-Mazo[2]

1) Hospital General Dr. Manuel Gea. Tlalpan 4800, 14080 Mexico City (México) 2) AB-Biotics SA (KANEKA Group), Torre Blanca 57, 08172 Barcelona (Spain)

PURPOSE

Gut microbiota has been reported to protect from lung viral infection in animal models by stimulating type-I interferon signaling.[1,2] Type-I interferons can have direct antiviral activity while also stimulating antibody-producing B cells.[3] Antibodies against Severe Acute Respiratory Syndrome coronavirus 2 (SARS-CoV2) have been correlated to faster infection clearance and protection against reinfection across viral variants.[4,5]

A 4-strain probiotic combination (AB21®) was recently studied in a randomized, placebo-controlled trial (ref. NCT04517422) in 300 Covid-19 ambulatory patients, whose infection had been confirmed by real-time polymerase chain reaction. Strains in probiotic were *Pediococcus acidilactici* CECT7483 (KABP021), *Lactiplantibacillus plantarum* CECT7484 (KABP022), *L. plantarum* CECT7485 (KABP023) and *L. plantarum* CECT30292 (KAPB033), at a total dose of 2x10^9cfu/day (verified at the end of the intervention period by standard plate counting). Study subjects did not receive corticosteroids or antivirals. Compared to placebo, probiotic intervention for 30 days achieved faster symptom clearance, a higher rate of viral clearance and increased SARS-CoV2-specific immunoglobulins M and G (IgM and IgG).[6] We thus hypothesize said probiotic effects could be related to increased type-I interferon signaling.

METHODS

- A random subset of 70 subjects (35 probiotic and 35 placebo) was selected out of the 300 participants in the clinical study. Stored serum samples collected on day 0 (baseline), 15 and 30 (end of intervention) were analyzed for interferon-alpha 2a (IFN-α) and interferon-beta 1a (IFN-β) using enzyme-linked immunosorbent assay. Demographic data, baseline viral load (as measured by real-time polymerase chain reaction on nasopharyngeal samples), symptom duration and SARS-CoV2-specific IgM and IgG serum titers were retrieved from patient case report forms.
- Differences between probiotic and placebo groups were compared using Mann-Whitney or Chi-squared tests, as appropriate. Correlations between type-I interferons and other variables were assessed by Spearman correlation.
- All procedures on study subjects had been approved by the Internal Review Board of Hospital General Dr. Manuel Gea (Mexico City) and adhered to Helsinki Declaration.

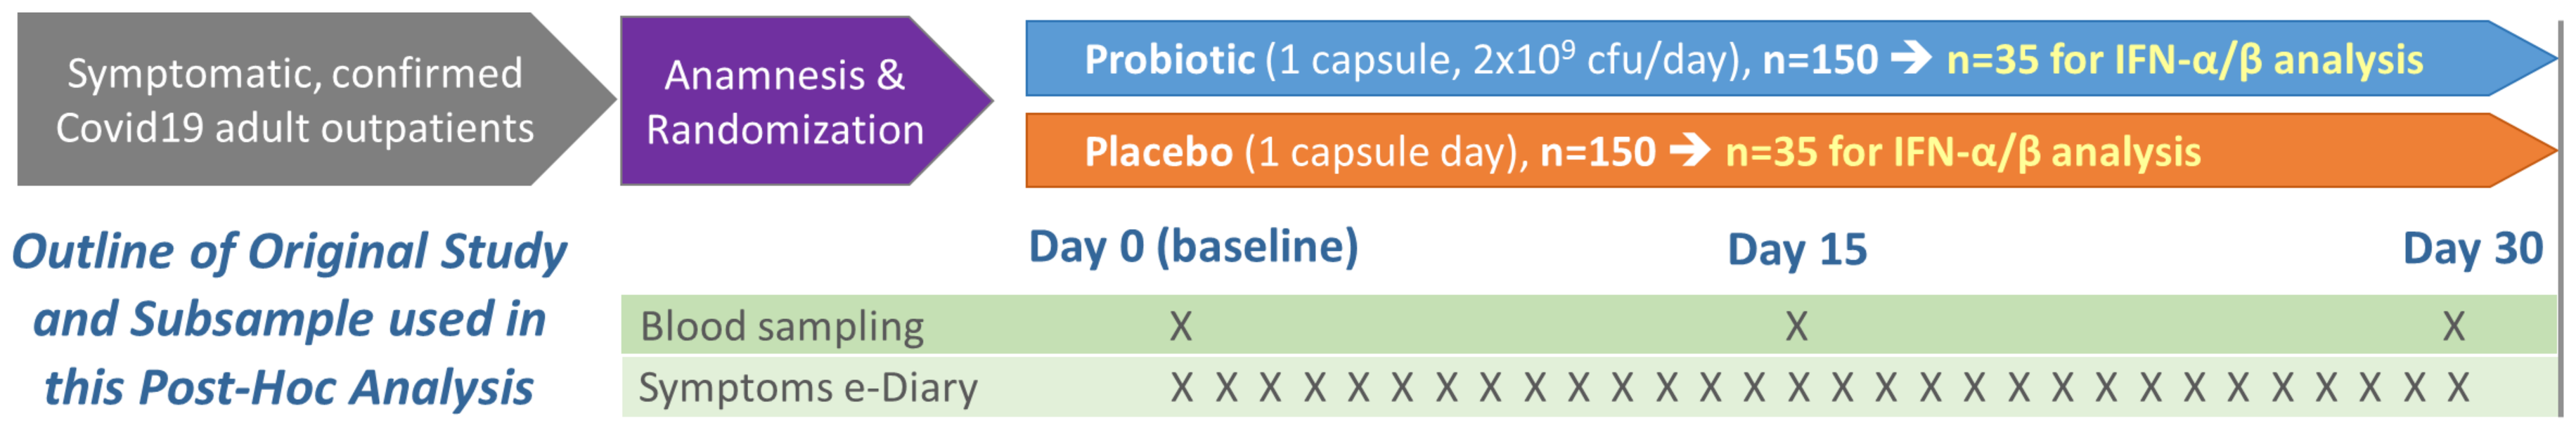
Symptomatic, confirmed Covid19 adult outpatients
Anamnesis & Randomization
Probiotic (1 capsule, 2x10⁹ cfu/day), n=150 → n=35 for IFN-α/β analysis
Placebo (1 capsule day), n=150 → n=35 for IFN-α/β analysis
Day 0 (baseline)
Day 15
Day 30
Blood sampling
Symptoms e-Diary

Outline of Original Study and Subsample used in this Post-Hoc Analysis

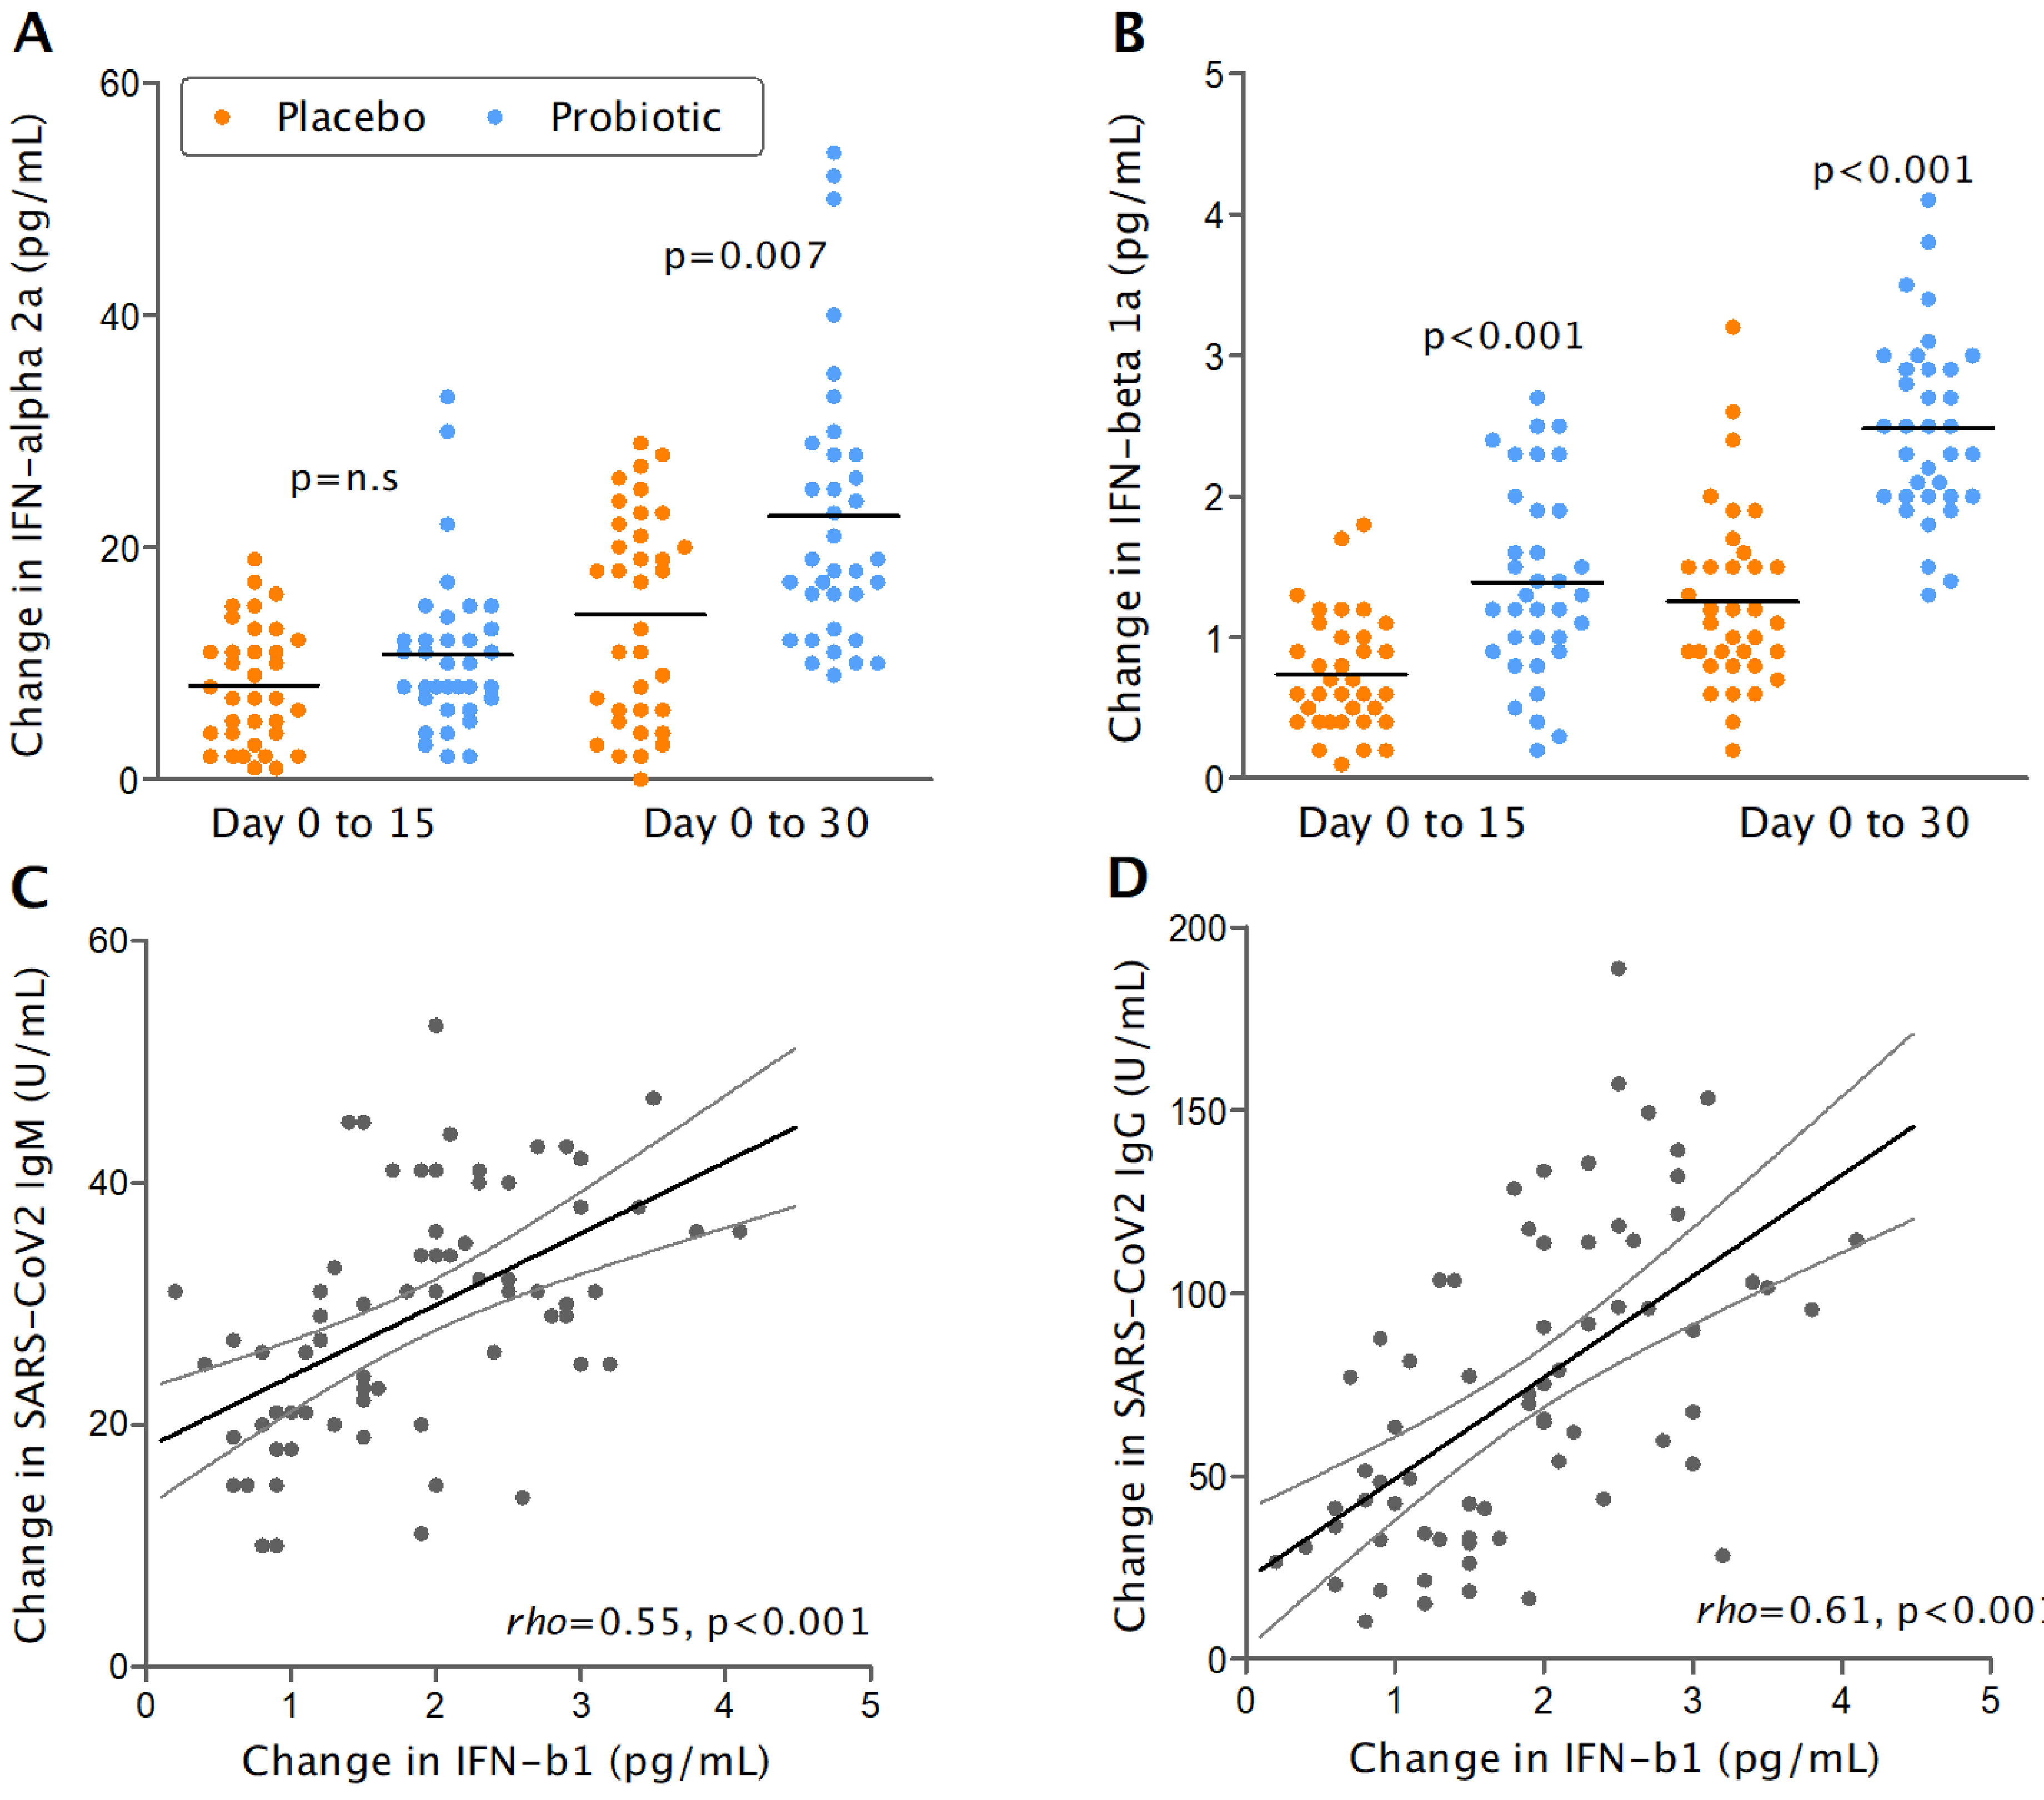
A
Placebo
Probiotic
Change in IFN-alpha 2a (pg/mL)
p=n.s
p=0.007
Day 0 to 15
Day 0 to 30
B
Change in IFN-beta 1a (pg/mL)
p<0.001
p<0.001
Day 0 to 15
Day 0 to 30
C
Change in SARS-CoV2 IgM (U/mL)
rho=0.55, p<0.001
Change in IFN-b1 (pg/mL)
D
Change in SARS-CoV2 IgG (U/mL)
rho=0.61, p<0.001
Change in IFN-b1 (pg/mL)

Baseline Characteristics of the Analyzed Population	Probiotic (n=35)	Placebo (n=35)
Age (years) [median, range]	34 (19-60)	40 (19-58)
Biological sex (female) [N, %]	22 (62.3%)	22 (62.3%)
Days from onset of symptoms [median, range]	4 (2-7)	4 (2-7)
SARS-CoV2 viral load (log copies/mL) [median, range][a]	6.80 [6.00-7.00]	6.75 [6.00-7.00]
Lung infiltrates (positive X-ray imaging) [N, %]	15 (42.9%)	11 (31.4%)
Body temperature (ºC) [median, range]	38.0 [37.2-39.1]	37.9 [36.9-39.1]
Peripheral oxygen saturation (SpO_2) < 92% [N, %][b]	26 (74.3%)	25 (71.4%)
Number of metabolic comorbidities [median, range][c]	0 [0-2]	0 [0-3]
Interferon-alpha 2a (pg/mL) [median, range]	12.0 [6.0-32-0]	11.0 [6.0-30.0]
Interferon-beta 1a (pg/mL) [median, range]	2.0 [0.9-3.0]	2.1 [1.1-2.7]
SARS-CoV2 IgM (arbitrary units) [median, range]	19.0 [8.0-22.0]	19.0 [11.0-22.0]
SARS-CoV2 IgG (arbitrary units) [median, range]	8.9 [0.3-32.4]	5.6 [0.9-31.3]

a) From high-sensitivity, real-time polymerase chain reaction of SARS-CoV2 *RdRp* gene (limit of detection: 0.01 viral copies/mL).[7]
b) All subjects had SpO2 ≥ 90% as per entry criteria. c) Comprising diabetes, arterial hypertension, and obesity (BMI ≥30 Kg/m^2)

RESULTS

- In the selected population for this post-hoc analysis, study groups were comparable at baseline (p>0.10 for all parameters, see Table).
- Duration of fever, cough, headache and myalgia were shorter (all p<0.05) in probiotic compared to placebo, averaging 2.5 to 3.3 less days, depending on symptom (not shown).
- Probiotic treatment resulted in larger increase of serum IFN-α on day 30 (p=0.007) and of IFN-β on days 15 and 30 (both p<0.001), compared to placebo (Figs. A,B, respectively).
- Similarly, SARS-CoV2-specific IgM and IgG were higher on days 15 and 30 (all p<0.001) in probiotic compared to placebo.
- Increase in IFN-β across the study correlated to increase in SARS-CoV2-specific IgM (*rho*=0.55, p<0.001, Fig.C) and IgG (*rho*=0.61, p<0.001, Fig. D), and inversely correlated to duration of cough (*rho*=-0.26, p=0.033) and fever (*rho*=-0.24, p=0.042).
- Increase in IFN-α across the study correlated to increase in SARS-CoV2-specific IgM only (*rho*=0.33, p=0.005) .
- Increases in IFN-α and IFN-β did not correlate to age, number of metabolic comorbidities, days from onset of symptoms or biological sex.

CONCLUSIONS

❖ Compared to placebo, oral intake of a specific 4-strain probiotic combination (AB21®) in Covid19 patients for 30 days, resulted in significant increase in type-I interferons in serum, especially IFN-β.

❖ Type-I interferons, especially IFN-β, correlated to higher SARS-CoV2-specific IgM and IgG antibodies and faster clearance of some symptoms.

❖ These observations suggest the AB21® probiotic formula may stimulate systemic, humoral antiviral response by inducing type-I interferons.

REFERENCES: 1) Abt et al. *Immunity* 2012, 37:158–170; 2) Bradley et al. *Cell Reports* 2019, 28:245–256; 3) Mc Nab et al. *Nat Rev Immunol.* 2015, 15:87-103; 4) Khoury et al. *Nat. Medicine* 2021, 27:1205-1211; 5) Cromer et al. *Lancet Microbe* 2022, 3:52-61; 6) Gutierrez-Castrellon et al. *Gut Microbes* 2022, 14:2018899; 7) WHO-Berlin Charité Protocol: Corman et al. *Euro Surveillance* 2020, 25:2000045

FUNDING: This study was wholly funded by KANEKA Corporation Ltd. (Japan).

HOSPITAL GENERAL DR. MANUEL GEA GONZALEZ

AB-BIOTICS Part of KANEKA Corporation

NO – PHOTO / RECORD